Glyphosate is Not an Endocrine Disruptor: Weight of Evidence versus Key Characteristic for Endocrine Disruption Assessment

D. Farmer[1], C. Borgert[2], D. Saltmiras[1], J. Klaunig[3], S. Levine[1], and J. DeSesso[4]. [1]Bayer Crop Science, Chesterfield, MO; [2]Applied Pharmacology and Toxicology Inc., Gainesville, FL; [3]Indiana University Bloomington, Bloomington, IN; and [4]Exponent, Alexandria, VA

APPLIED PHARMACOLOGY AND TOXICOLOGY, INC.
Consulting & Research Services

INTRODUCTION

1

Regulatory authorities (US EPA 2015, EFSA 2017, Assessment Group on Glyphosate (AGG/Rapporteur Member States for EU Annex Renewal) 2022) and a peer reviewed publication (Levine et al., 2020) have used the established weight of evidence (WoE) approach and evaluated other scientifically relevant information from validated test systems and publications as the basis for their endocrine disruption (ED) assessments on glyphosate (GLY)

2

These WoE assessments have consistently concluded that GLY lacks ED properties due to its lack of potential to act via estrogen, androgen, thyroid or steroidogenic (EATS) modes of action (MoAs)

3

In contrast, a publication by Munoz et al. (2020) concluded the opposite based on a novel set of 10 Key Characteristics (KCs) of endocrine-disrupting chemicals (EDCs) developed by La Merrill et al. (2020)

- Le Merrill et al:
 - devised an approach that has not been empirically tested or validated
 - derived their concept for the approach based on flawed analogies to a similar approach developed for carcinogens and ignored analyses revealing its poor performance
 - mischaracterized their proposal as a consensus recommendation, when in fact, they followed no recognized method for developing a consensus document and failed to include experts who have published contrasting interpretations
 - ignored fundamental principles of hormone action that have been known for more than a century and well-established characteristics of dose-response in endocrine pharmacology and toxicology
 - developed an approach that lacks a means to reach a negative conclusion about a chemicals' ED properties

4

Munoz et al. found GLY to possess 8 of the 10 KCs proposed by La Merrill et al. for ED (see red circles). To understand this striking discrepancy, we compared the WoE and the KC approach against several established regulatory evaluation criteria (US EPA, 2020, EFSA, 2017, OECD, 2019). The authors:

- omitted regulatory guideline studies including those on GLY's reproductive and developmental toxicity and endocrine disrupting potential and numerous relevant and reliable publications
- relied on a small subset of results from non-standard and unvalidated *in vitro* and *in vivo* assays that are not traditionally used to inform regulatory safety assessments
- did not apply, a means of determining the relevance, reliability and quality of data selected for their assessment
- did not address conflicting results
- did not consider the basic principles of pharmacology, hormone action, and dose-response
- selected 50 publications of which almost half included studies that tested GLY-based formulations (GBF) and the results reported in these studies result from non-endocrine effects (e.g. cytotoxicity)

5

As examples, two tables provide a list of the publications Munoz et al. used to support their conclusion of GLY meeting the KCs of KC 1. ("current evidence indicates that glyphosate can favor hormonal receptor activity, particularly ERα by stimulating their transcriptional activation and therefore promoting phenotype changes in breast cell line models. Nevertheless, the molecular mechanism of interaction is unknown.") and for evidence that glyphosate had effects on different hormone-producing or hormone-sensitive cells as depicted in their Figure 1. Also provided in the tables are the test materials, study type, route of exposure and results/comments on the listed publications

1 Regulatory WoE Scientific Assessment

UNITED STATES ENVIRONMENTAL PROTECTION AGENCY

OECD BETTER POLICIES FOR BETTER LIVES

efsa European Food Safety Authority

Problem formulation
- Define the question(s) for assessment

- Evidence collection (establish lines of evidence and knowledge gaps)
- Evidence evaluation (determine data reliability, uncertainty and relevance)
- Evidence weighing (assign weight to evidence)
- Evidence integration and reporting (examine evidence coherence)

Uncertainty analysis
- Assess and combine uncertainties from all parts of the overall assessment
- Identify data gaps

Conclusion of overall assessment

2 US EDSP Tier 1 Assays

Assays	Test Guideline
ER binding (Rat uterine cytosol)	OSCPP 890.1250
ERα Transcriptional Activation Assay (Human cell line HeLa 9903)	OSCPP 890.1300; OECD 455
AR Binding Assay (Rat prostate cytosol)	OSCPP 890.1150
Steroidogenesis Assay (Human cell line H295R)	OSCPP 890.1550; OECD 456
Aromatase Assay (human recombinant microsomes)	OSCPP 890.1200
Uterotrophic Assay (Rat)	OSCPP 890.1600; OECD 440
Hershberger Assay (Rat)	OSCPP 890.1400; OECD 441
Pubertal Female Assay (Rat)	OSCPP 890.1450
Pubertal Male Assay (Rat)	OSCPP 890.1500
Fish Short-term Reproduction Assay	OSCPP 890.1350; OECD 229
Amphibian Metamorphosis Assay (Frog)	OSCPP 890.1100; OECD 231

Other Scientifically Relevant Information

Study Type	Number of Studies	Test Guidelines	Endpoints
In Silico			Estrogen receptor, androgen receptor, thyroid receptor, glycocorticoid receptor, mineralocorticoid receptor, liver X receptor, peroxisome proliferator-activated receptor, retinoid X receptor, aryl hydrocarbon receptor, pregnane X receptor, and CYP3A4 activation
Subchronic Toxicity	31	OPPTS 870.3050, 870.3100, 870.3150, 870.3200	Growth, ovarian weight, adrenal weight, testes weight and histopathological evaluation of the following organs and/or systems – uterus, ovary, thyroid, adrenal, pituitary, oviduct, parathyroid, epididymides, testis, prostate and seminal vesicle
Chronic Toxicity/ Carcinogenicity	27	OPPTS 870.4200, 870.4300	Growth, ovarian weight, thyroid weight, adrenal weight, testes weight, histopathological evaluation of the following organs and/or systems - uterus, ovary, thyroid, adrenal, pituitary, epididymides, testis, prostate, seminal vesicle, and preputial gland
Developmental Toxicity	15	OPPTS 870.3700	Growth, uterine weight, pup malformations
Reproductive Toxicity	9	OPPTS 870.3800	In addition to reproductive parameters, intersex and sex ratio these endpoints are examined in animals exposed during critical developmental stages including prenatal, early postnatal and the peripubescent period into adulthood Females: ovarian weight, uterine weight, adrenal weight, pituitary weight, thyroid weight, growth, age at vaginal opening, time to mating, estrus, implantation, in utero development, lactation, histopathological evaluation of the following organs and/or systems – uterus, ovary, thyroid, adrenal, pituitary and oviduct Males: testes weight, adrenal weight, thyroid weight, pituitary weight, epididymides weight, accessory sex organ weight, growth, age at preputial separation, histopathological evaluation of the following organs and/or systems – epididymides, testes, thyroid, adrenal, pituitary, prostate and seminal vesicle
Publications	23	None	Numerous

AGG (2022) Glyphosate ED Assessment for Humans: "In conclusion, glyphosate does not induce EATS-mediated adversity and no EATS-related endocrine activity was observed in silico, in vitro, and in vivo."

Which Assessment Do You Trust?

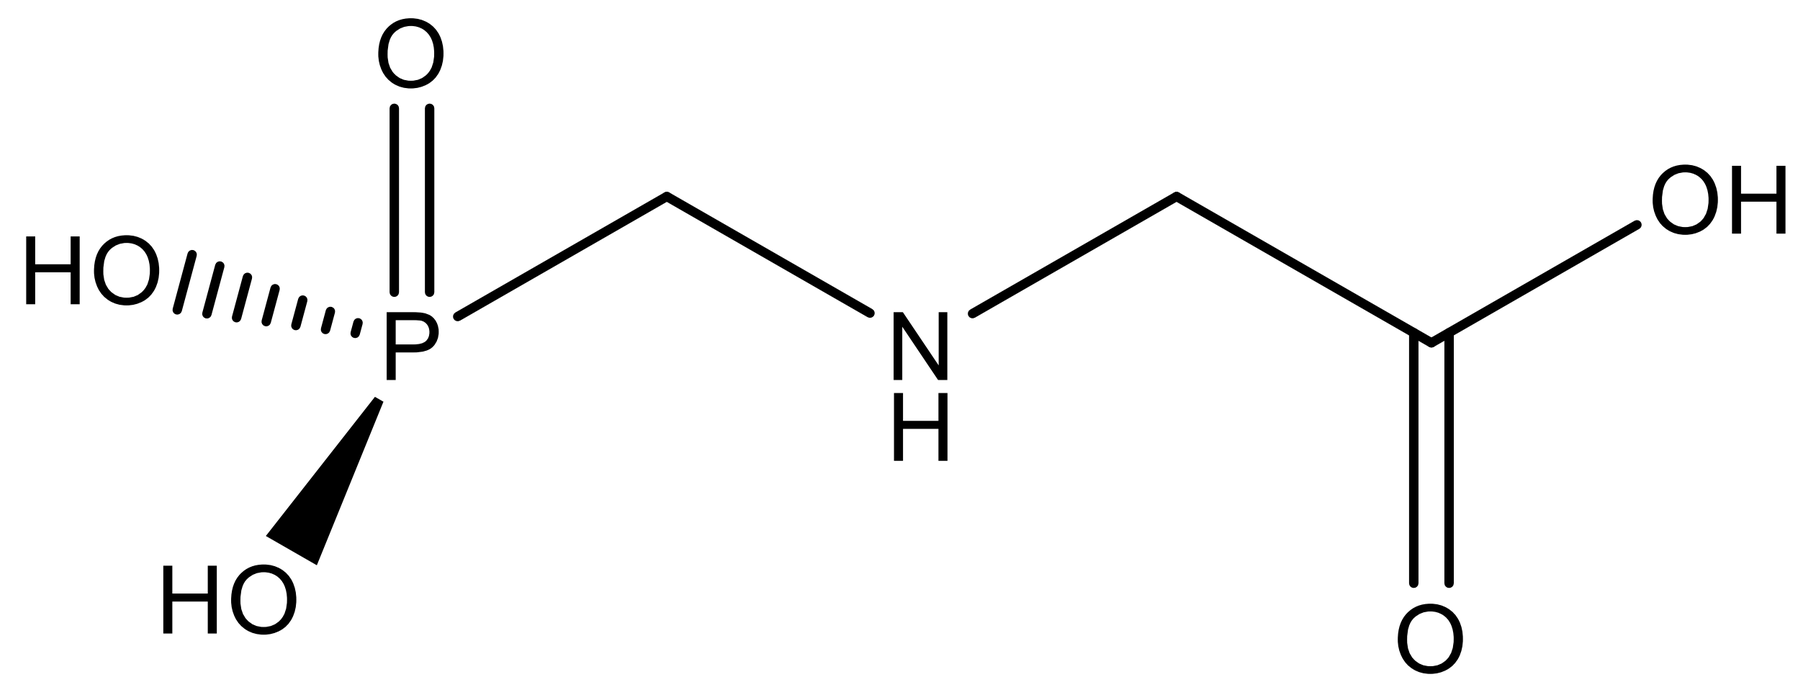

Glyphosate (N-phosphonomethyl) glycine

Which Results Do You Believe?

CONCLUSIONS

- **Regulatory WoE assessments are clear that GLY does not show EATS modalities.**
- **The methodological deficiencies of KCs explain the incorrect classification of glyphosate's potential as an ED using this novel approach compared to the application of well-established WoE approaches developed by regulatory agencies over the past decade.**
- **ED KCs like other KCs have not been validated against negative controls and the GLY data is an ideal example of a negative control and demonstrates how the KC approach lacks a means to get to a negative conclusion about a chemical's ED properties.**

Key Characteristics

3

NATURE REVIEWS | ENDOCRINOLOGY VOLUME 16 | JANUARY 2020 | 45

CONSENSUS STATEMENT

OPEN

Consensus on the key characteristics of endocrine-disrupting chemicals as a basis for hazard identification

Michele A. La Merrill[1]*, Laura N. Vandenberg[2], Martyn T. Smith[3], William Goodson[4], Patience Browne[5], Heather B. Patisaul[6], Kathryn Z. Guyton[7], Andreas Kortenkamp[8], Vincent J. Cogliano[9], Tracey J. Woodruff[10], Linda Rieswijk[3,11], Hideko Sone[12], Kenneth S. Korach[13], Andrea C. Gore[14], Lauren Zeise[15] and R. Thomas Zoeller[16]

4

Chemosphere 270 (2021) 128619

Contents lists available at ScienceDirect

Chemosphere

journal homepage: www.elsevier.com/locate/chemosphere

ELSEVIER

Review

Glyphosate and the key characteristics of an endocrine disruptor: A review

Juan P. Muñoz [a], Tammy C. Bleak [a], Gloria M. Calaf [a,b,*]

[a] Instituto de Alta Investigación (IAI), Universidad de Tarapacá, Antofagasta 1520, Arica, 1000000, Chile
[b] Center for Radiological Research, VC11-218, Columbia University Medical Center, 630 West 168th Street, New York, NY, 10032, USA

Table 1 | Key characteristics of EDCs and representative standardized tests that address them

Key characteristics	Examples of relevant streams of mechanistic evidence	Guideline description (species) [agency and guideline number]*
KC1. Interacts with or activates hormone receptors	Binding or agonism of hormone receptors	Androgen Receptor Binding (rat) [US EPA 890.1150]; Estrogen Receptor Binding (rat) [US EPA 890.1250, OECD TG 493]; Estrogen Receptor Transcriptional Activation (human stable transfection) [US EPA 890.1300, OECD TG 455]; Androgen Receptor Binding (rat) [US EPA 890.1150]; Androgen Receptor Transcriptional Activation (human stable transfection) [OECD TG 458]; Uterotrophic (rat) [US EPA 890.1600, OECD TG 440]; Hershberger [US EPA 890.1400, OECD TG 441]
KC2. Antagonizes hormone receptors	Antagonism of nuclear or cell surface hormone receptors	Estrogen Receptor Transcriptional Activation (human) [OECD TG 455]; Androgen Receptor Transcriptional Activation (human) [OECD TG 458]; Hershberger [US EPA 890.1400, OECD TG 441]
KC3. Alters hormone receptor expression	Abundance, distribution and degradation of hormone receptors	None
KC4. Alters signal transduction in hormone-responsive cells	Abundance of post-translational modifications, cofactors, transcription factors and transcripts, and activity of associated enzymes	None
KC5. Induces epigenetic modifications in hormone-producing or hormone-responsive cells	Chromatin modifications, DNA methylation and non-coding RNA expression	None
KC6. Alters hormone synthesis	Expression or activity of enzymes or substrates in hormone synthesis	Aromatase (human) [US EPA 890.1200]; Steroidogenesis (human) [US EPA 890.1550, OECD TG 456]
KC7. Alters hormone transport across cell membranes	Intracellular transport, vesicle dynamics or cellular secretion	None
KC8. Alters hormone distribution or circulating hormone levels	Blood protein expression and binding capacity, blood levels of pro-hormones and hormones	None
KC9. Alters hormone metabolism or clearance	Inactivation, breakdown, recycling, clearance, excretion or elimination of hormones	None
KC10. Alters fate of hormone-producing or hormone-responsive cells	Atrophy, hyperplasia, hypertrophy, differentiation, migration, proliferation or apoptosis	None

EDC, endocrine-disrupting chemical; OECD, Organisation for Economic Co-operation and Development; TG, test guideline; US EPA, US Environmental Protection Agency. *Only assays that serve as the basis of regulatory decisions of the OECD and US EPA are provided.

This table was copied from the publication of La Merrill et al. and modified to indicate the 8 KCs that Munoz et al. concluded that glyphosate satisfied

5

Publications cited by Munoz et al to support their conclusion that GLY meets the criteria for KC 1. Interacts with or activates hormone receptors

Publication	Test Material	Type of Study—All In Vitro	Results/Comments
Kojima et al. 2004	GLY	Chinese hamster ovary (CHO-K1) cells that were transiently transfected with an estrogen responsive reporter gene	Findings consistent with the US EDSP and EFSA evaluations
Hokanson et al. 2007	GBF	MCF-7 is a human breast cancer cell line	GBF not applicable for evaluation of GLY
Gasnier et al. 2009	GLY/GBF	ERα, ERβ transcriptional activities in human liver cancer HepG2 cells that were transiently transfected (ERE-TK-Luciferase) and AR measured in MDA-MB453-kb2 cells	GBF not applicable for evaluation of GLY; findings consistent with the US EDSP and EFSA evaluations for anti-estrogenic activity but not consistent with aromatase activity
Thongprakaisang et al. 2013	GLY	Hormone-dependent human breast cancer, T47D, a stably ERE-luc construct transfected hormone-dependent breast cancer, T47D-KBluc, and a hormone-independent human breast cancer, MDA-MB231	Findings are not consistent with the US EPA EDSP; authors report GLY had a greater estrogenic effect than a maximally inducing dose of 17 β-estradiol 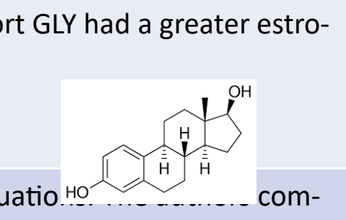
Mesnage et al. 2017	GLY	Hormone-dependent (MCF-7 and T47D) and hormone-independent (MDA-MB-231) human breast cancer cell lines as well as T47D-Kluc (which stably expresses an estrogen receptor transactivation assay (ERTA) reporter construct)	Finding are not consistent with the US EPA EDSP and EFSA evaluations. The authors comment that the activation of ERα occurred at relatively high concentrations (1000 to 100,000 μ/L compared 17 β-estradiol at 0.0013 μ/L) through a ligand-independent pathway and that activation would not occur at typical exposure levels in human and commented that the findings of Thongprakaisang et al. were possibly the result of contamination

Publications cited by Munoz et al as depicted in Figure 1. of their publication as a "Summary of evidences related to the effects of glyphosate and its derivatives on different hormone-producing or hormone-sensitive cells"

Publication	Test Material	Type of Study	Route of Exposure	Results/Comments
Altamirano et al. 2018	GBF	*In Vivo*	S.C Injection	GBF not applicable for evaluation of GLY; route of exposure not relevant for human exposure
De Liz Oliveria et al. 2013	GBF	*In Vitro*		GBF not applicable for evaluation of GLY
De Souza et al. 2017	GBF	*In Vivo*	Oral Gavage	GBF not applicable for evaluation of GLY
Duforestel et al. 2019	GLY	*In Vitro*		Findings are inconsistent with US EDSP and EFSA evaluations
Hokanson et al. 2007	GBF	*In Vitro*		GBF not applicable for evaluation of GLY
Guerrero et al. 2017	GBF	*In Vivo*	S.C. Injection	GBF not applicable for evaluation of GLY; route of exposure not relevant for human exposure
Lorenz et al. 2019	GBF	*In Vivo*	Oral Dietary	GBF not applicable for evaluation of GLY
Mesnage et al. 2017	GLY/GBF	*In Vitro*		GBF not applicable for evaluation of GLY; GLY findings not consistent with US EDSP and EFSA evaluations
Richard et al. 2005	GLY/GBF	*In Vitro*		GBF not applicable for evaluation of GLY; GLY findings not consistent with US EDSP and EFSA evaluations; inhibition was observed only at concentrations of GLY that were cytotoxic
Sritana et al. 2018	GLY	*In Vitro*		Findings not consistent with US EDSP and EFSA evaluations; possible estrogenic contamination
Stur et al 2019	GBF	*In Vitro*		GBF not applicable for evaluation of GLY
Thongprakaisang et al. 2013	GLY	*In Vitro*		Findings not consistent with US EDSP and EFSA evaluations; possible estrogenic contamination
Walsh et al. 2000	GLY/GBF	*In Vitro*		GBF not applicable for evaluation of GLY; GLY findings were consistent with US EDSP and EFSA evaluations